RECCE® 327 Demonstrates Bactericidal Activity against Several Microbial Species

Authors: Michele Dilizia[1], Hannah Tsunemoto[2], Diana Quach[2], Marc Sharp[2], John Prendergast[1], James Graham[1]

1 WHO. *Antimicrobial Resistance*. World Health Organization; 2019. Accessed July 1, 2022. https://www.who.int/news-room/fact-sheets/detail/antimicrobial-resistance.
2 Dadgostar P. Antimicrobial Resistance: Implications and Costs. *IDR*. 2019;Volume 12:3903-3910. doi:10.2147/IDR.S234610.
3 Jonas O, Irwin A, Berthe F, Cesar Jean L, Gall FG, Marquez PV. *DRUG-RESISTANT INFECTIONS: A Threat to Our Economic Future*. World Bank Group; 2017:1-172.

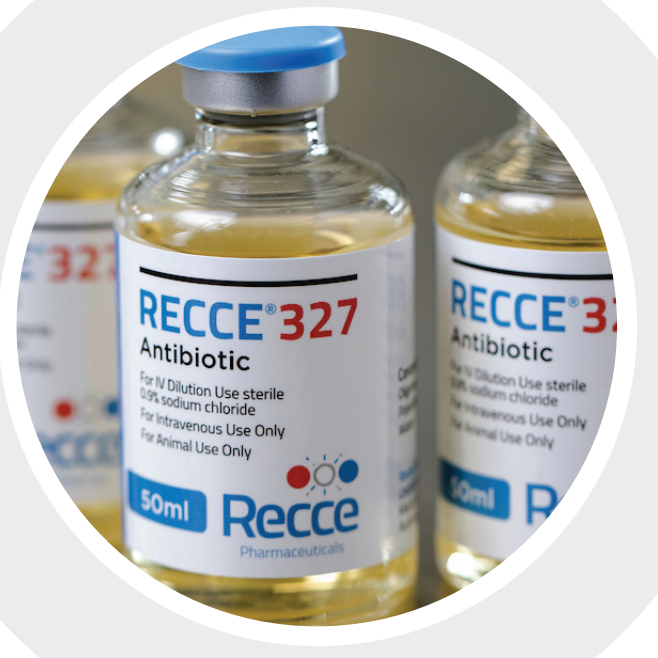
RECCE®327
Antibiotic
50ml
Recce

Background

The World Health Organization has recognized the growing threat of antimicrobial resistance (AMR) as one of the top 10 dangers to humanity.[1] Common infections are becoming increasingly resistant to available therapies resulting in multi-resistant and pan-resistant microbial species, including *Enterococcus faecium, Staphylococcus aureus, Klebsiella pneumoniae, Acinetobacter baumannii, Pseudomonas aeruginosa* and *Enterobacter* better known as ESKAPE pathogens.[2,3] Beyond AMR's impact on human health, it poses significant economic costs, leading to longer hospital stays, the need for more expensive medicines and a greater financial burden on patients.[2,3] We must take appropriate action to develop antimicrobial substances that not only kill resistant pathogens but also circumvent the mechanisms of resistance.

Materials

We investigated the activity of a new synthetic polymer anti-infective with rapid and potent broad-spectrum bactericidal activity known as RECCE® 327 (R327) against Gram-positive, Gram-negative, and mycobacterial species, including the ESKAPE pathogens, *B. subtilis* and in biofilms of *E. coli* to determine the effects of R327 on these bacterial species.

Results

Our results demonstrate that R327 is rapidly bactericidal, reducing viable cell counts across all tested bacterial species and conditions. Cells treated with R327 showed rapid, dose dependent, decreases in cellular ATP levels in luciferase based *in-vitro* ATP assays. R327 treatment of the mycobacteria *M. avium* and *M. abscessus* also led to a decline in ATP levels, though more slowly likely due to the low growth rate of these strains. In some species of gram-positive bacteria, the *in-vitro* ATP assay indicated transient increases in ATP levels upon treatment with R327 at high concentrations, while viable cell counts showed rapid cell death under the same conditions.

Conclusion

These data suggest that the apparent increase in ATP levels may be an artifact of the in vitro kit and imply that the MOA of R327 may involve destabilization of the cell envelope. Our results further reinforce the broad-spectrum bactericidal activity of R327 and demonstrate its potential as a new anti-infective.

E. coli — RECCE® 327 activity in 10% LB: *E. coli* ATCC 25922

ATP Levels in *E. coli* ATCC 25922 Treated with RECCE® 327 in 10% LB

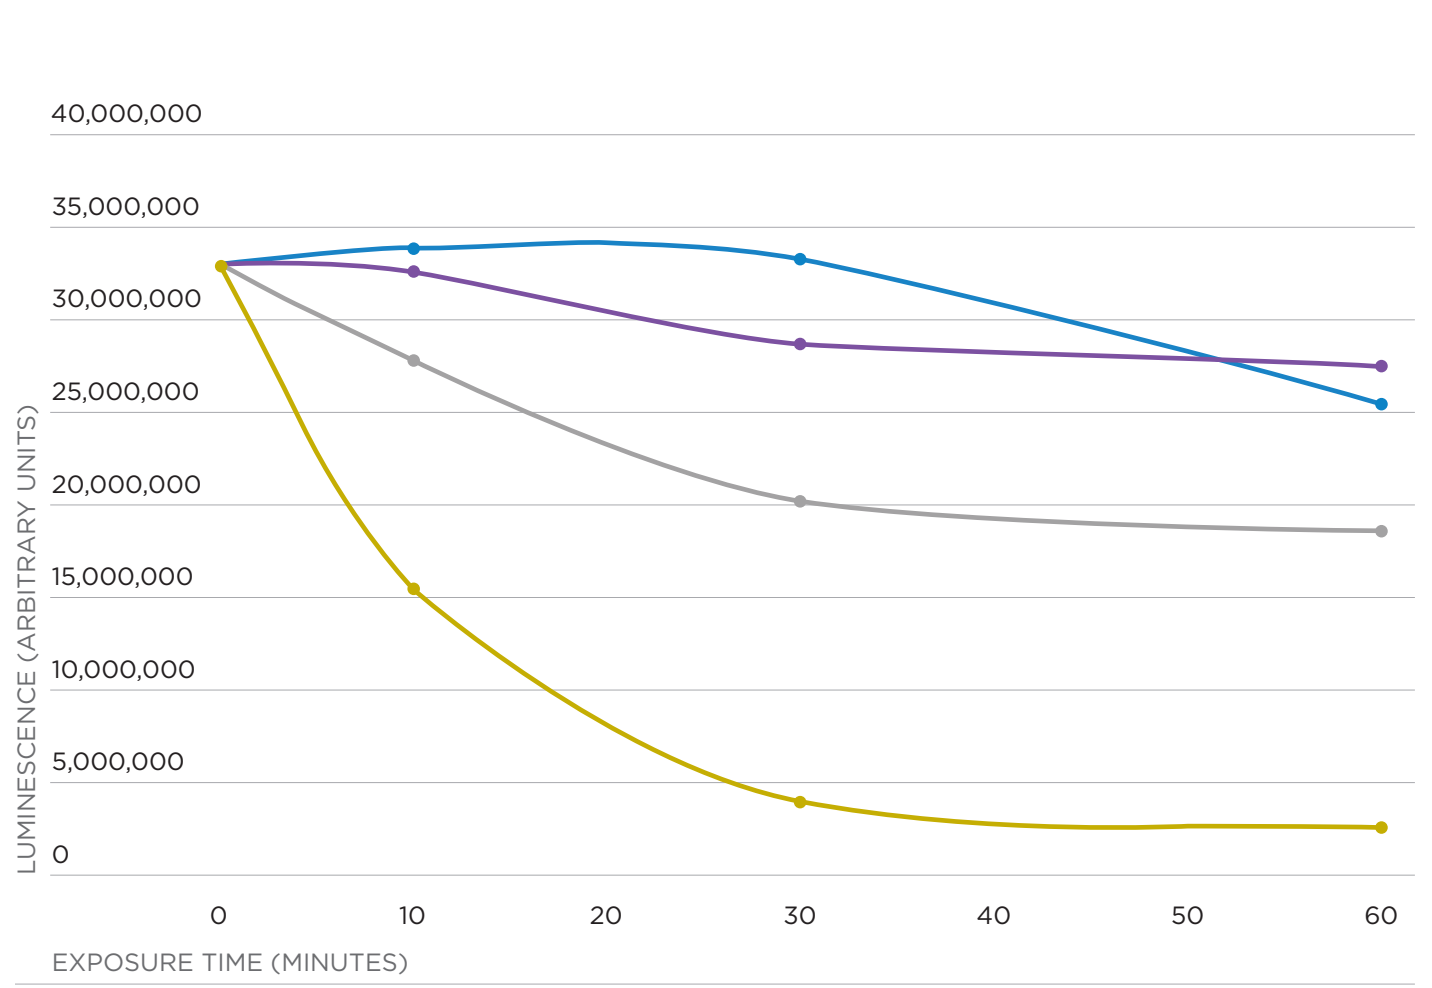
LUMINESCENCE (ARBITRARY UNITS)
40,000,000
35,000,000
30,000,000
25,000,000
20,000,000
15,000,000
10,000,000
5,000,000
0
0 10 20 30 40 50 60
EXPOSURE TIME (MINUTES)

Viable Cells in *E. coli* ATCC 25922 Treated with RECCE® 327 in 10% LB

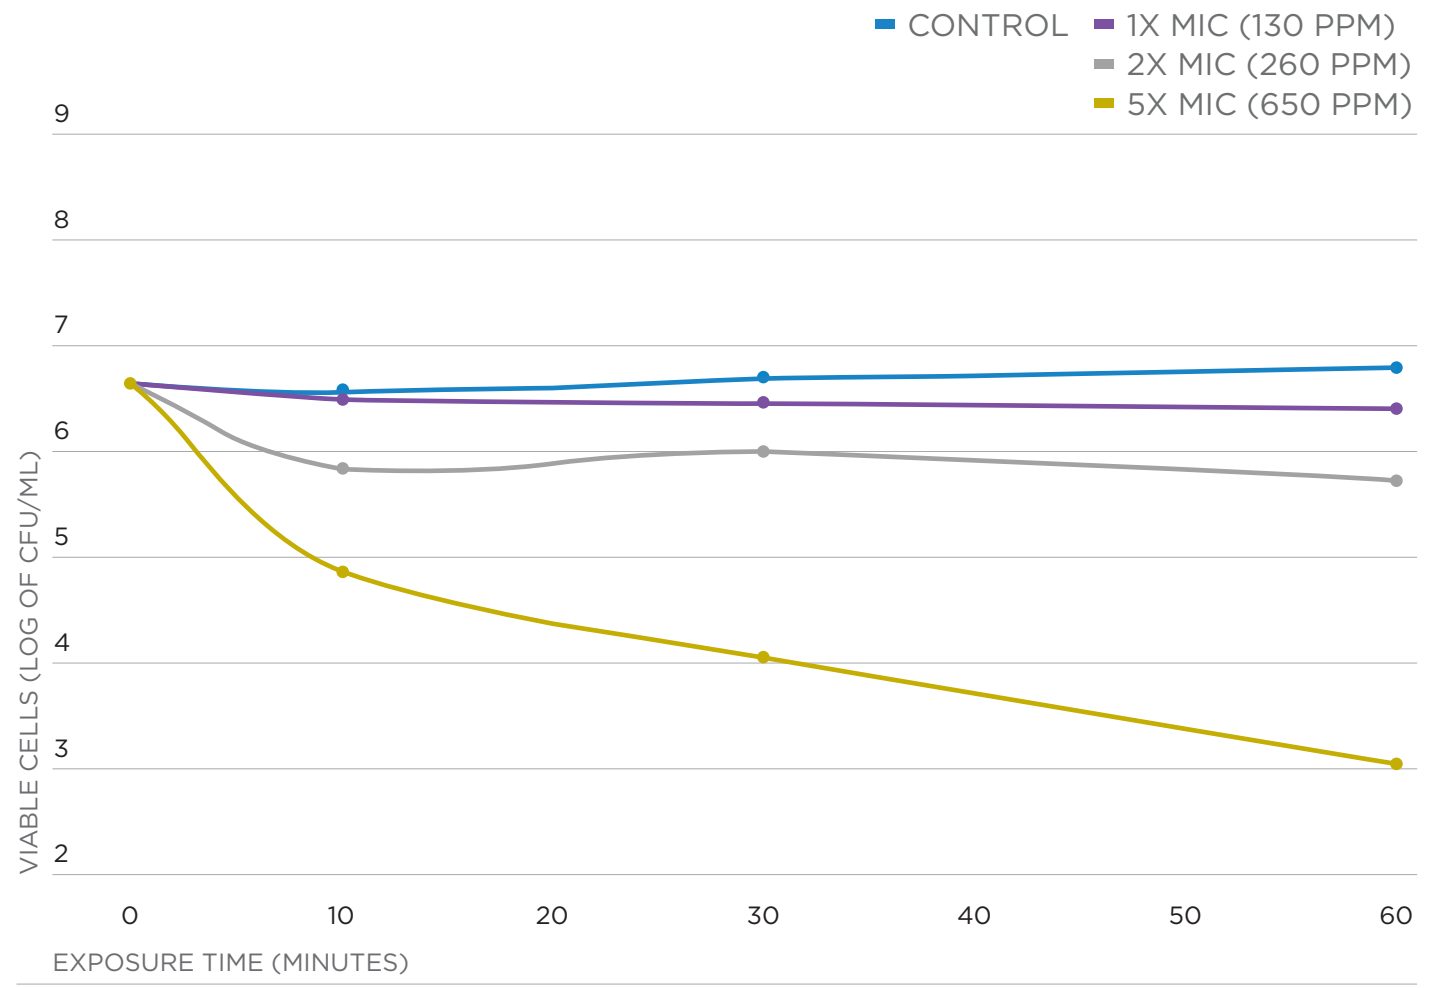
CONTROL
1X MIC (130 PPM)
2X MIC (260 PPM)
5X MIC (650 PPM)
VIABLE CELLS (LOG OF CFU/ML)
9 8 7 6 5 4 3 2
0 10 20 30 40 50 60
EXPOSURE TIME (MINUTES)

B. subtilis — RECCE® 327 activity in 10% LB: *B. subtilis* PY79

ATP Levels in *B. subtilis* PY79 Treated with RECCE® 327 in 10% LB

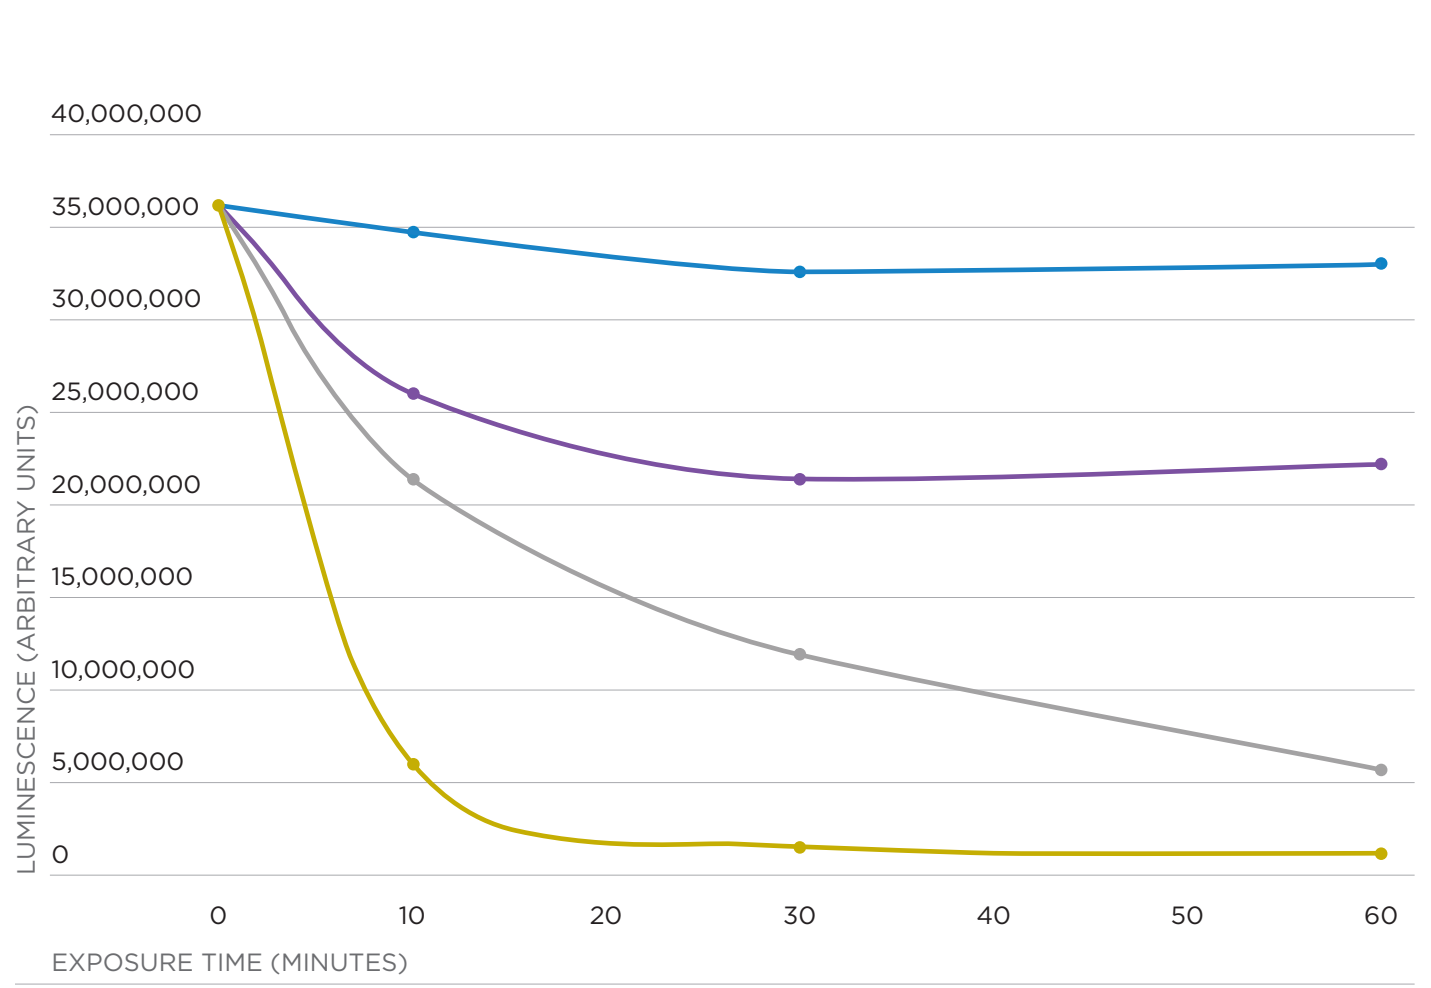
LUMINESCENCE (ARBITRARY UNITS)
40,000,000
35,000,000
30,000,000
25,000,000
20,000,000
15,000,000
10,000,000
5,000,000
0
0 10 20 30 40 50 60
EXPOSURE TIME (MINUTES)

Viable Cells in *B. subtilis* PY79 Treated with RECCE® 327 in 10% LB

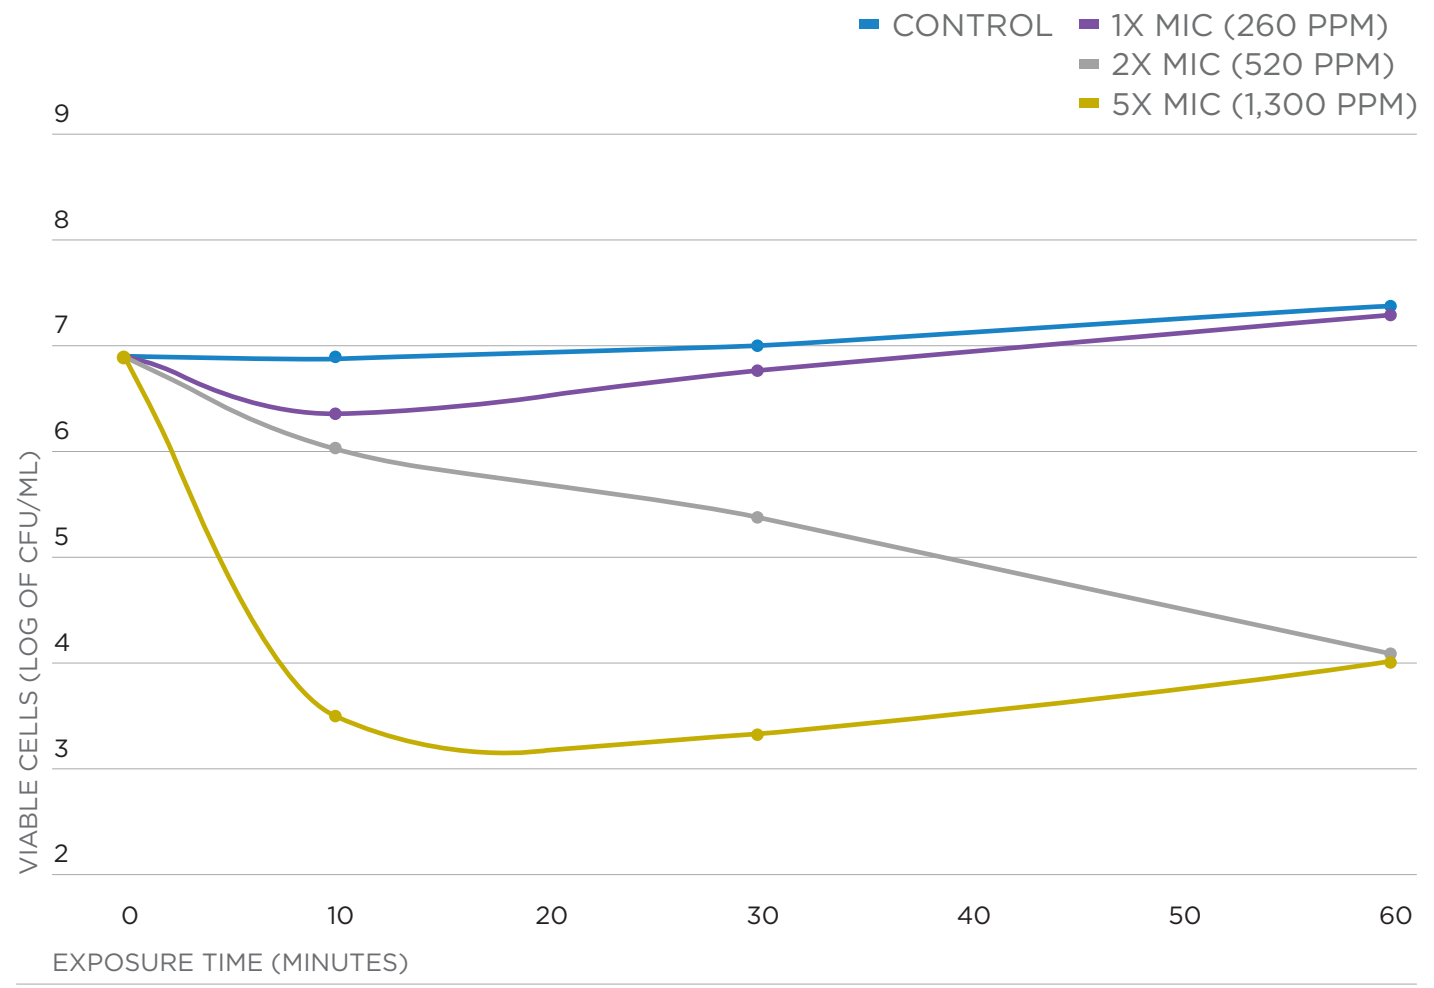
CONTROL
1X MIC (260 PPM)
2X MIC (520 PPM)
5X MIC (1,300 PPM)
VIABLE CELLS (LOG OF CFU/ML)
9 8 7 6 5 4 3 2
0 10 20 30 40 50 60
EXPOSURE TIME (MINUTES)

P. aeruginosa — RECCE® 327 activity in 10% LB: *P. aeruginosa* PAO1

ATP Levels in *P. aeruginosa* PAO1 Treated with RECCE® 327 in 10% LB

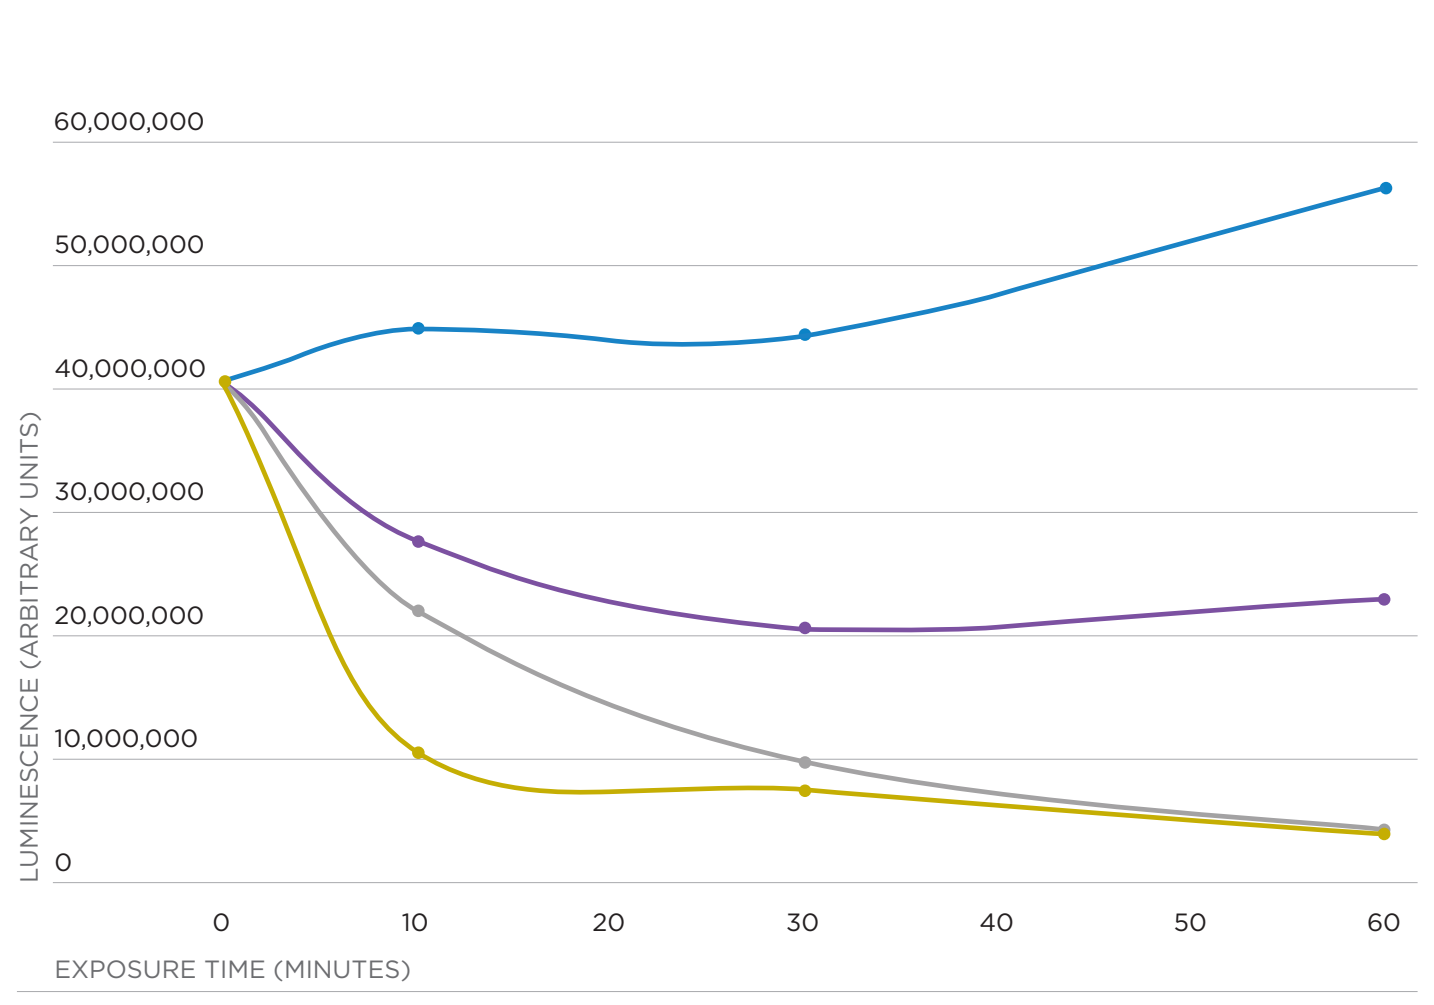
LUMINESCENCE (ARBITRARY UNITS)
60,000,000
50,000,000
40,000,000
30,000,000
20,000,000
10,000,000
0
0 10 20 30 40 50 60
EXPOSURE TIME (MINUTES)

Viable Cells in *P. aeruginosa* PAO1 Treated with RECCE® 327 in 10% LB

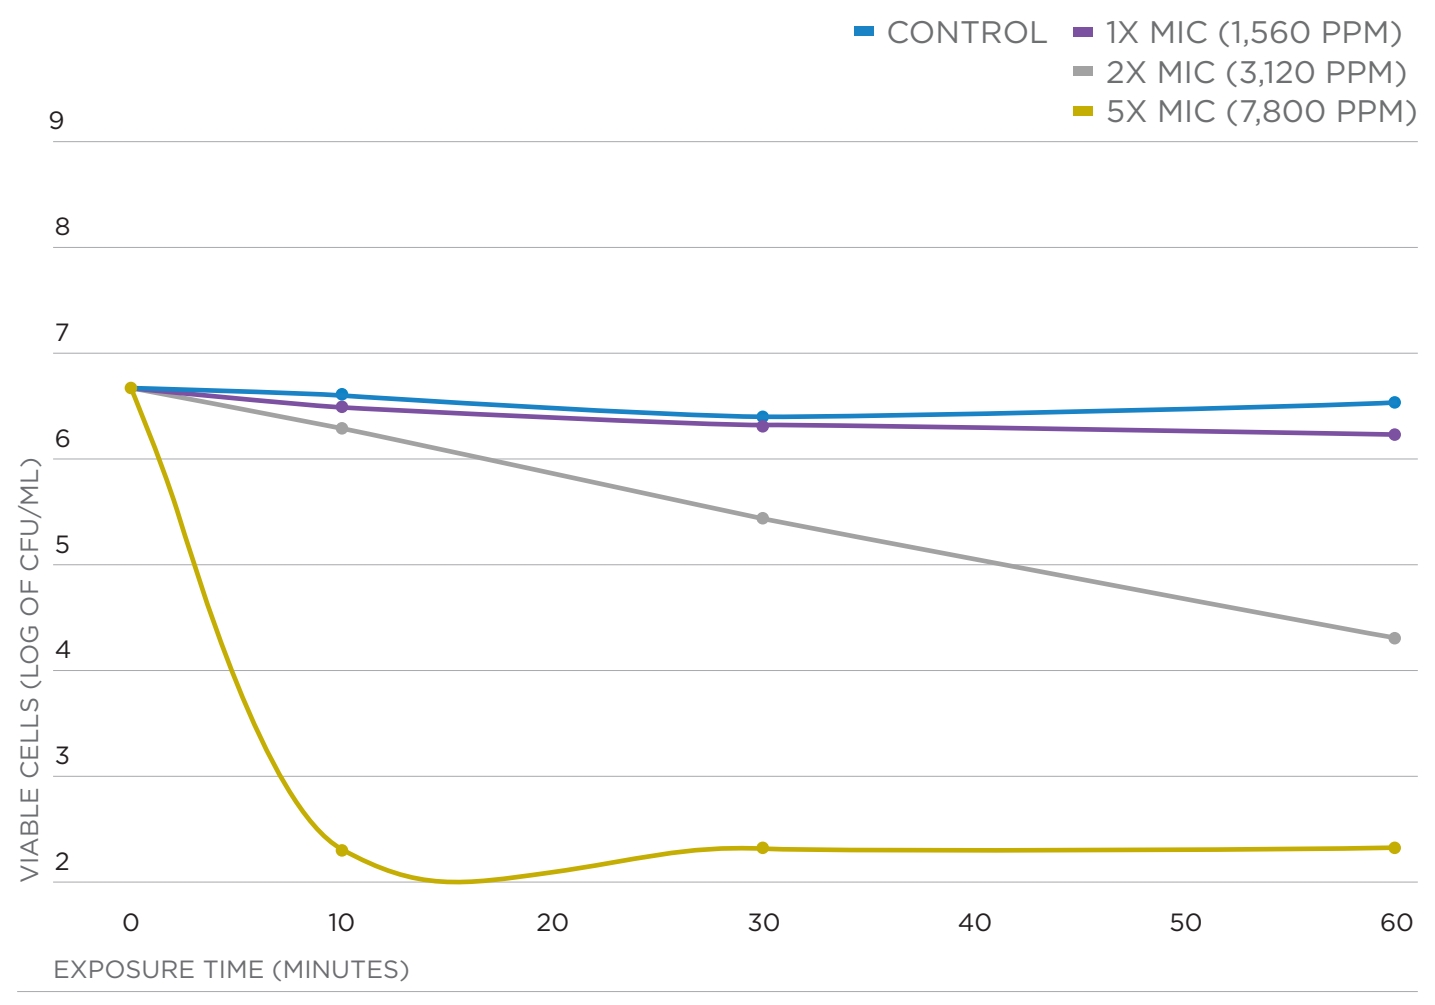
CONTROL
1X MIC (1,560 PPM)
2X MIC (3,120 PPM)
5X MIC (7,800 PPM)
VIABLE CELLS (LOG OF CFU/ML)
9 8 7 6 5 4 3 2
0 10 20 30 40 50 60
EXPOSURE TIME (MINUTES)

A. baumannii — RECCE® 327 activity in 10% LB: *A. baumannii* ATCC 17978

ATP Levels in *A. baumannii* ATCC 17978 Treated with RECCE® 327 in 10% LB

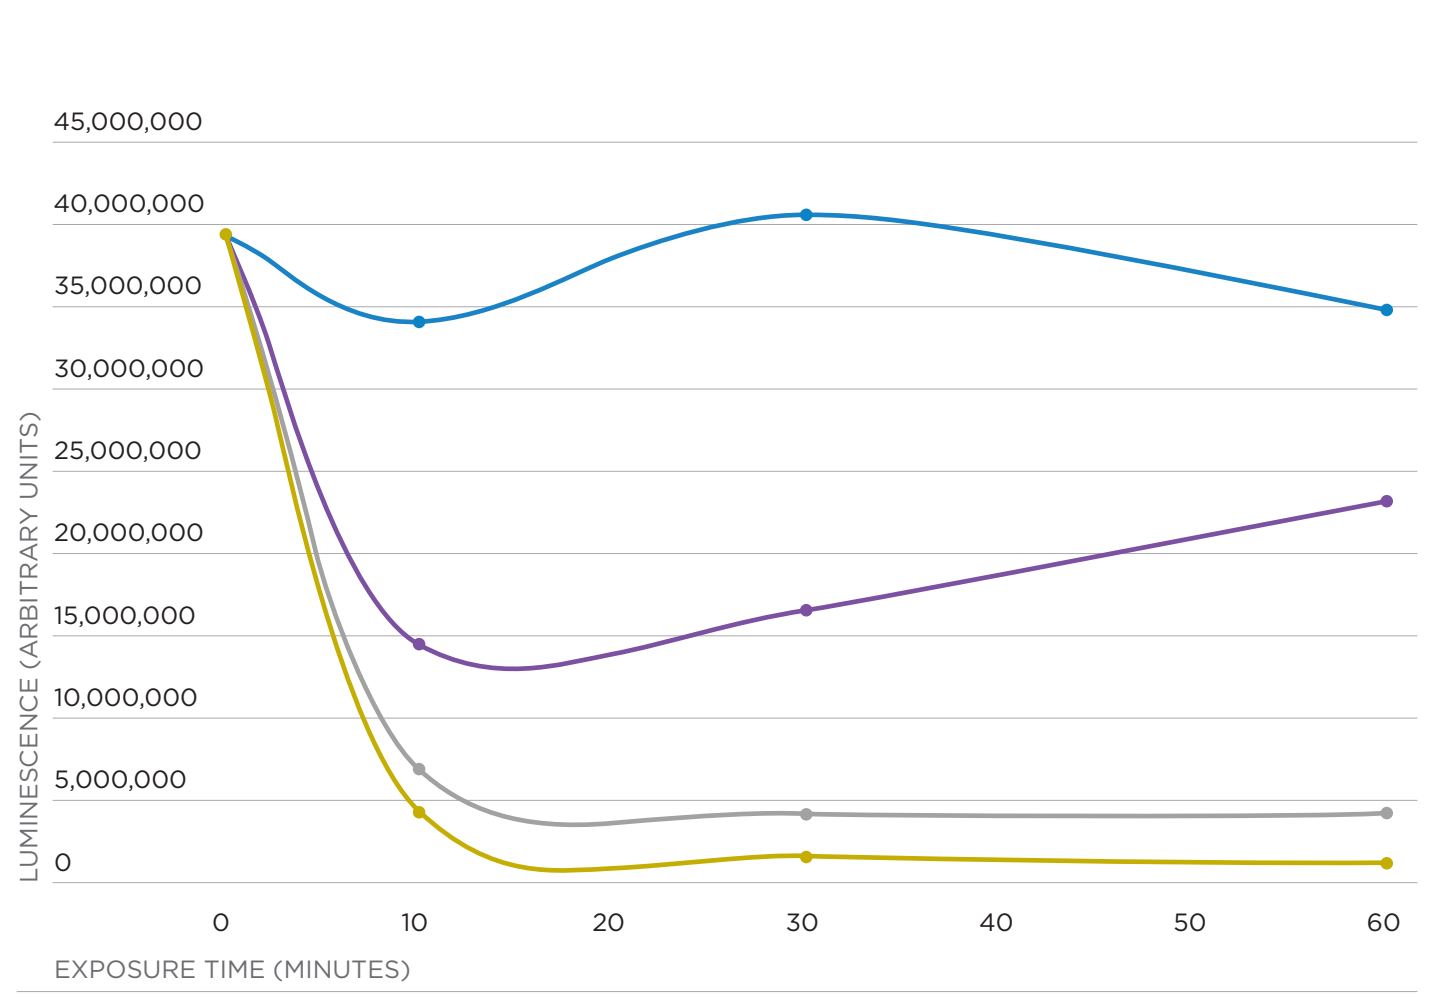
LUMINESCENCE (ARBITRARY UNITS)
45,000,000
40,000,000
35,000,000
30,000,000
25,000,000
20,000,000
15,000,000
10,000,000
5,000,000
0
0 10 20 30 40 50 60
EXPOSURE TIME (MINUTES)

Viable Cells in *A. baumannii* ATCC 17978 Treated with RECCE® 327 in 10% LB

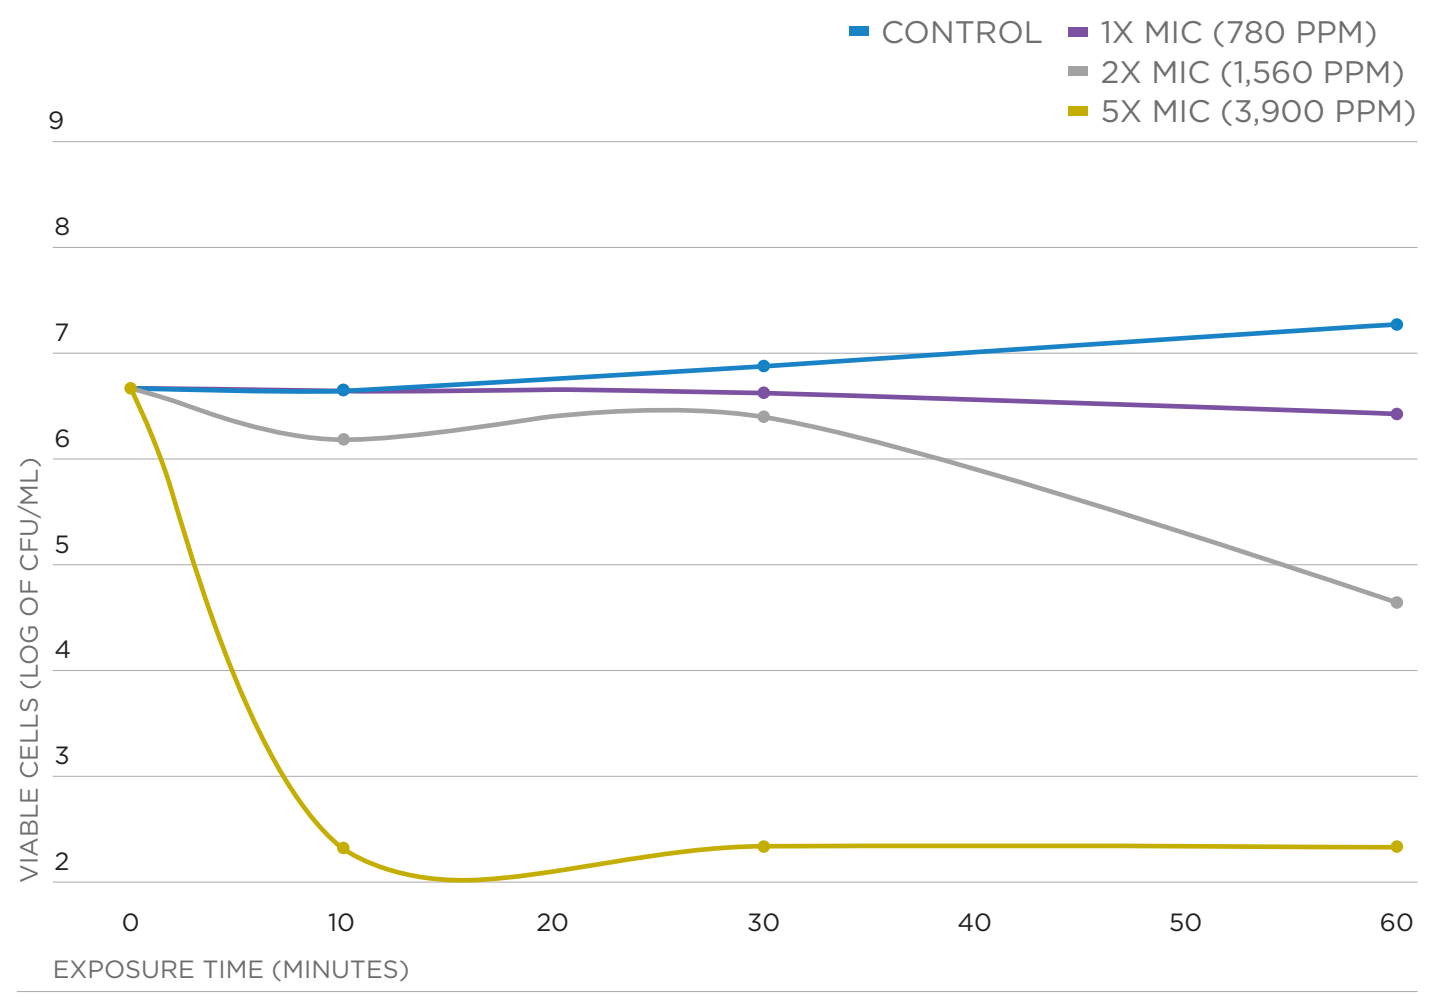
CONTROL
1X MIC (780 PPM)
2X MIC (1,560 PPM)
5X MIC (3,900 PPM)
VIABLE CELLS (LOG OF CFU/ML)
9 8 7 6 5 4 3 2
0 10 20 30 40 50 60
EXPOSURE TIME (MINUTES)

K. pneumoniae — RECCE® 327 activity in 10% LB: *K. pneumoniae* ATCC 700603

ATP Levels in *K. pneumoniae* ATCC 700603 Treated with RECCE® 327 in 10% LB

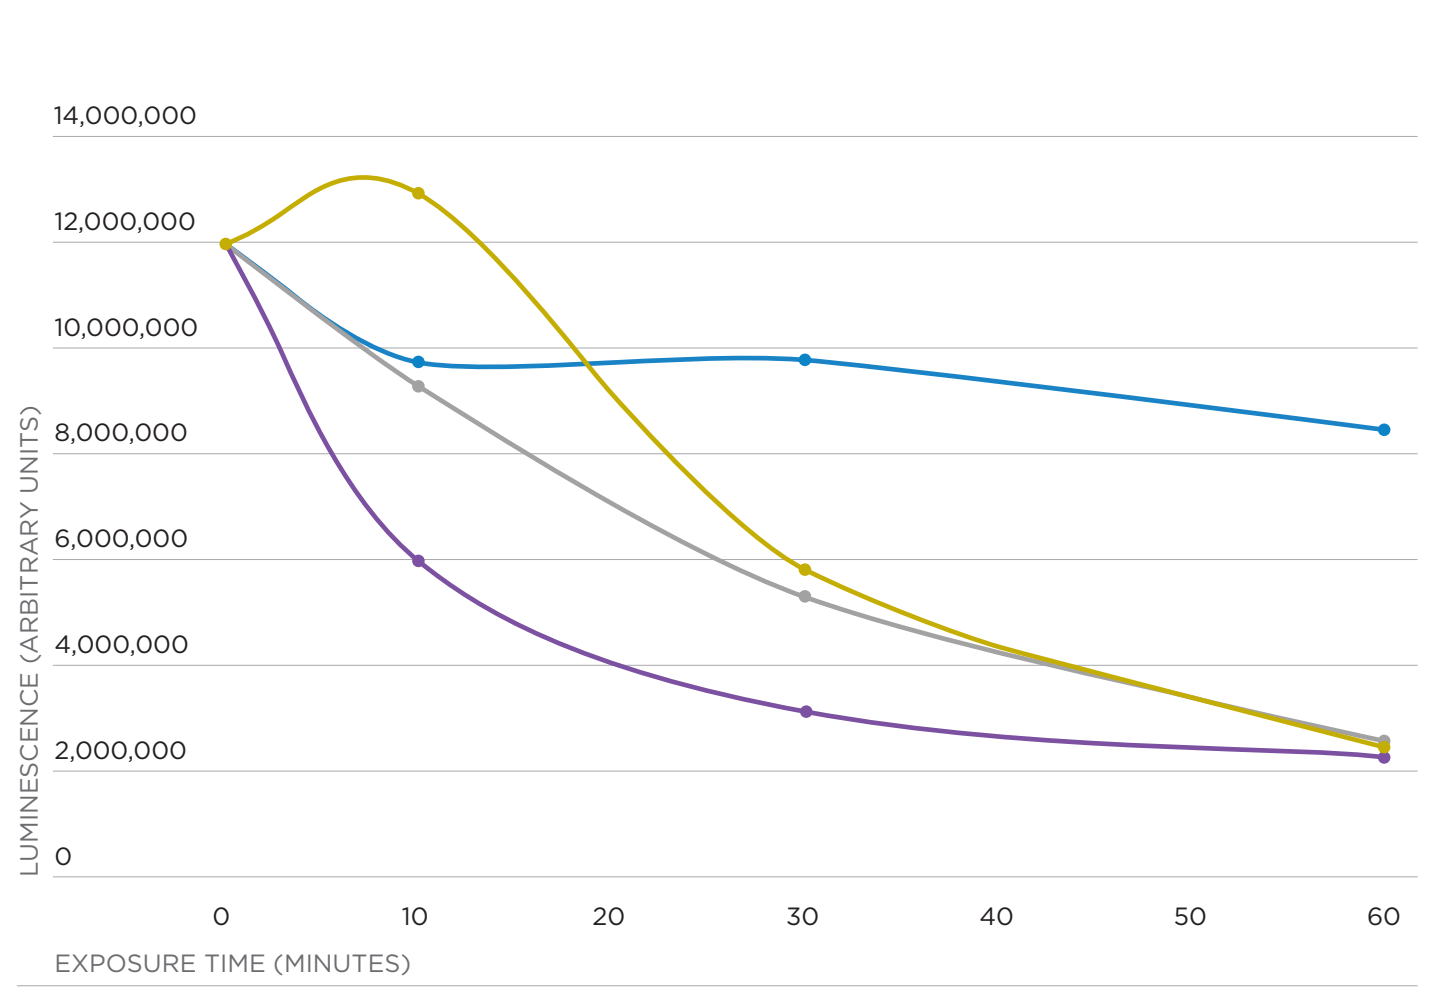
LUMINESCENCE (ARBITRARY UNITS)
14,000,000
12,000,000
10,000,000
8,000,000
6,000,000
4,000,000
2,000,000
0
0 10 20 30 40 50 60
EXPOSURE TIME (MINUTES)

Viable Cells in *K. pneumoniae* ATCC 700603 Treated with RECCE® 327 in 10% LB

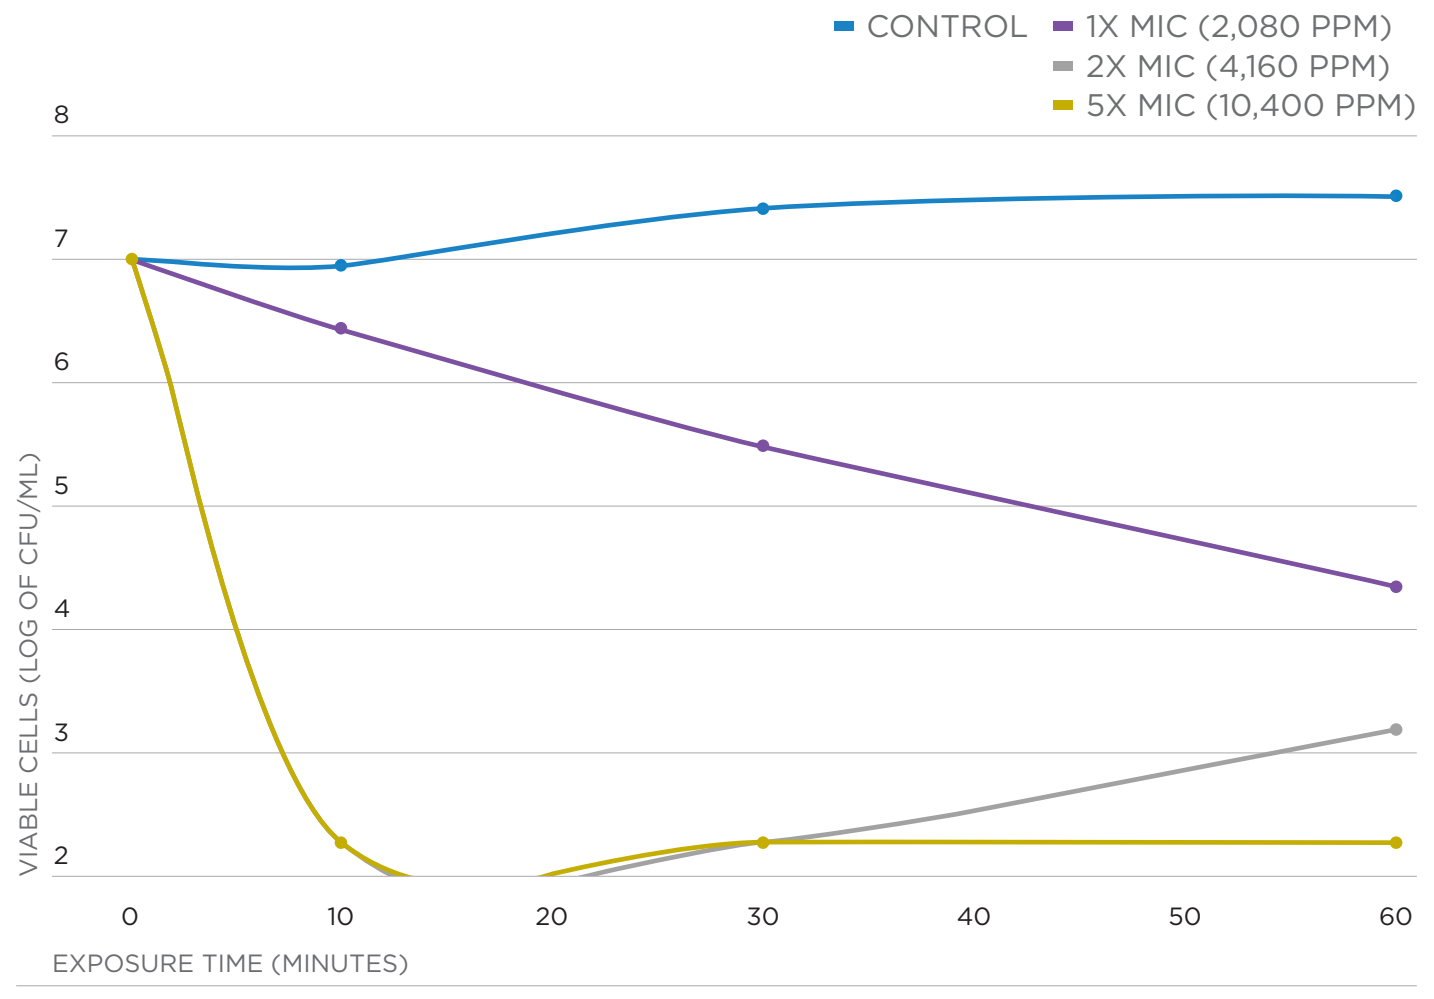
CONTROL
1X MIC (2,080 PPM)
2X MIC (4,160 PPM)
5X MIC (10,400 PPM)
VIABLE CELLS (LOG OF CFU/ML)
8 7 6 5 4 3 2
0 10 20 30 40 50 60
EXPOSURE TIME (MINUTES)

S. aureus — RECCE® 327 activity in 10% LB: *S. aureus* ATCC 29213

ATP Levels in *S. aureus* ATCC 29213 Treated with RECCE® 327 in 10% LB

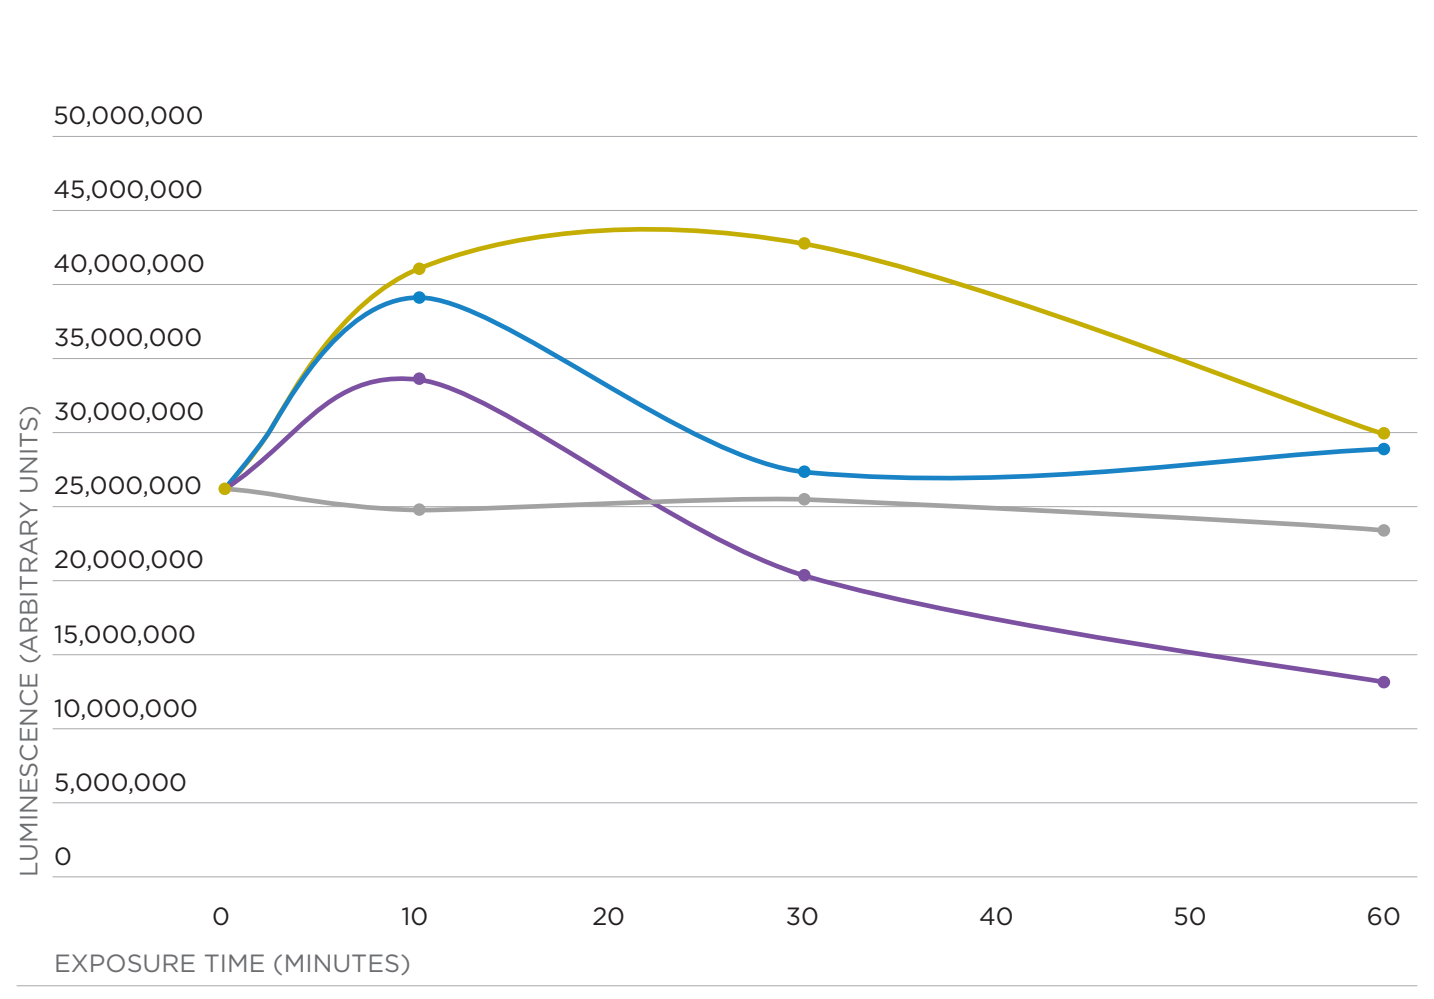
LUMINESCENCE (ARBITRARY UNITS)
50,000,000
45,000,000
40,000,000
35,000,000
30,000,000
25,000,000
20,000,000
15,000,000
10,000,000
5,000,000
0
0 10 20 30 40 50 60
EXPOSURE TIME (MINUTES)

Viable Cells in *S. aureus* ATCC 29213 Treated with RECCE® 327 in 10% LB

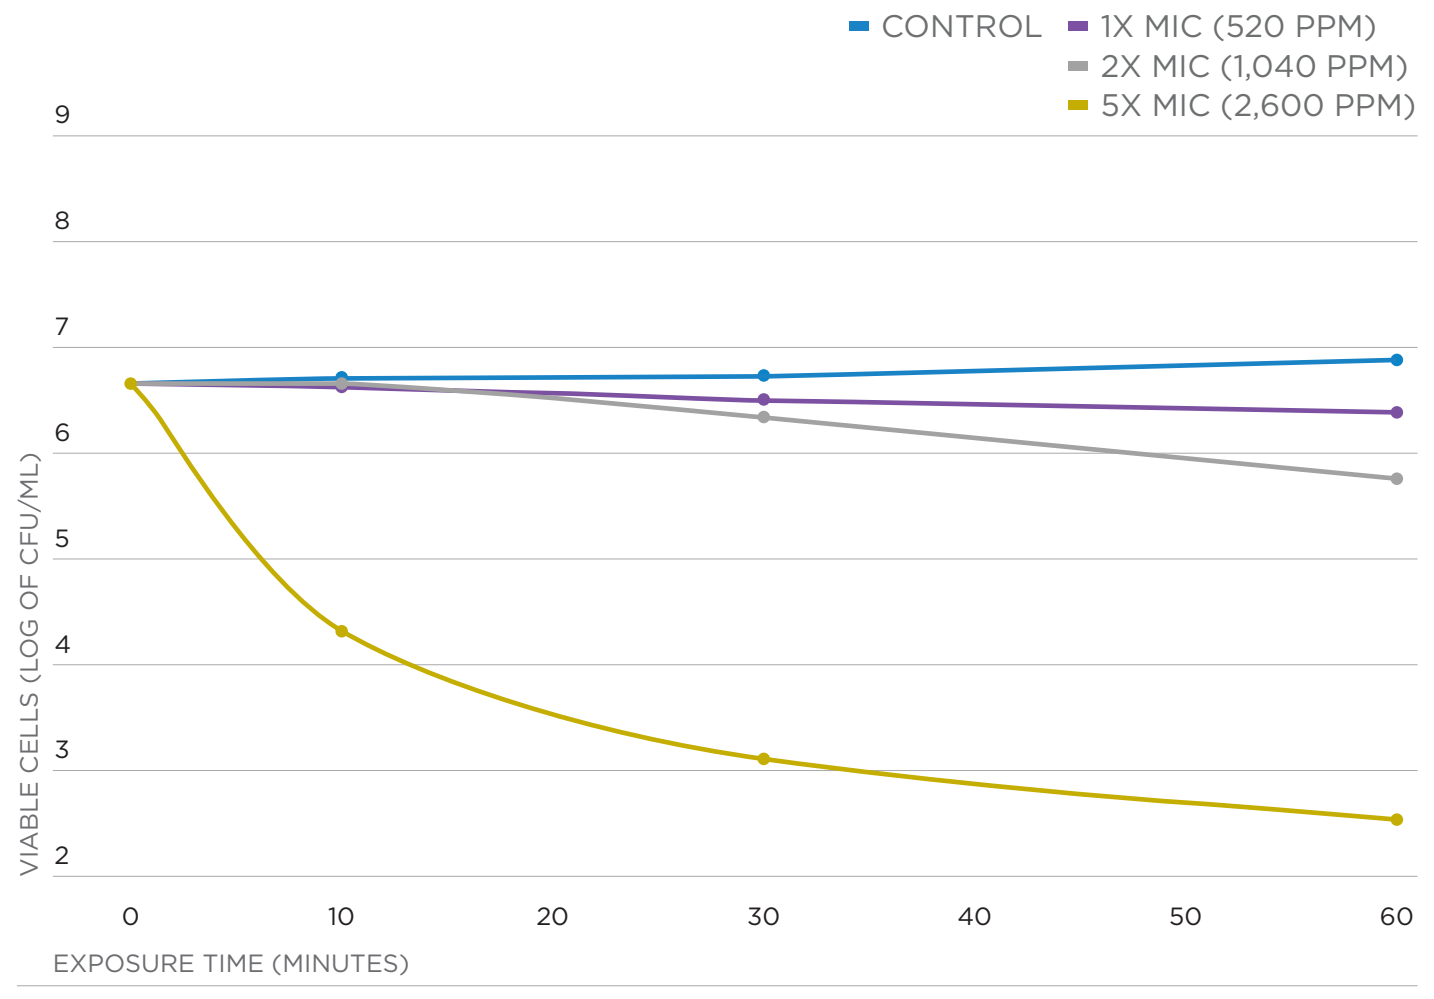
CONTROL
1X MIC (520 PPM)
2X MIC (1,040 PPM)
5X MIC (2,600 PPM)
VIABLE CELLS (LOG OF CFU/ML)
9 8 7 6 5 4 3 2
0 10 20 30 40 50 60
EXPOSURE TIME (MINUTES)

Recce Pharmaceuticals

recce.com.au
ASX:RCE FSE: R9Q